Characterization of patient perception of odor severity in hidradenitis suppurativa using TARGET-DERM HS

Tiffany Mayo MD [1], Jayesh Patel PhD [2], Bethanee J Schlosser MD, PhD [2], Beth Rycroft PhD [2], Amy E Gamelli PhD [2], Claire C Bristow PhD, MPH, MSc [2], Akash Danavar PharmD, MS, MPH [2], Feng Yu PhD[3], Benjamin H Kaffenberger MD [4], Michael Cecchini MD [5], Alexa Kimball MD, MPH[6] on behalf of TARGET-DERM HS Investigators

1. University of Alabama Birmingham, Alabama 2. AbbVie Inc., Illinois 3. Target RWE, North Carolina 4. Ohio State University, Ohio 5. York Dermatology Clinic and Research Centre, Canada 6. Harvard Medical School, Massachusetts

TARGET RWE
BETTER EVIDENCE • BETTER HEALTH

Introduction

- HS-related odor (or smell) is frequently reported by HS patients but rarely quantitatively evaluated from a patient perspective.
- It is unclear if differences exist across the continuum of odor severity and which demographic and/or disease characteristics may be associated with greater odor severity.
- The aim of this patient-centric analysis is to identify characteristics associated with HS odor severity in a real-world HS patient cohort.

Methods

- This is a cross-sectional analysis of HS patients ≥12 years who completed the Hidradenitis Suppurativa Severity Assessment (HSSA) [1] at enrollment into TARGET-DERM HS, a longitudinal observational registry in the US and Canada.[2]
- Without a specific cutoff to define severe HS-related odor, we categorized patients into distinct quartiles based on their perceived odor severity, quartiles were derived from patient responses to the HSSA Question #8 (Odor Severity).
- Descriptive statistics were calculated including means, medians, frequency and distributions of the clinician- and patient-reported outcomes (listed below).
- Characteristics were compared across all quartiles using Jonckheere-Terpstra, Cochran-Armitage or Cochran-Mantel-Haenszel, Kruskal-Wallis, Chi-squared, or Fisher's exact test where appropriate.
- Multivariable modelling for HiSQoL, DLQI, PROMIS-Depression, and PROMIS-Anxiety was completed using HSSA Odor Severity question, demographics, and clinician-reported outcomes as covariates.

Clinician-Reported Outcomes at Enrollment

- HS-PGA, Hidradenitis Suppurative Physician Global Assessment, 0 (Clear) – 5 (Very severe)
- Hurley Stage, 1 – 3
- The anatomical locations (body sites included abdomen, axillae, breast, buttock, groin, thigh, and other) and the numerical counts of abscesses, nodules, fistulas, and the sum of abscesses and nodules.

Patient-Reported Outcomes:

- HSSA Question 8 Odor Severity, "In the past 7 days, how bad was the smell coming from the area(s) affected by your HS? 0 (No bad smell) – 10 (Extremely bad smell)
- HiSQoL, Hidradenitis Suppurativa Quality of Life 0 (No effect) – 68 (Extreme effect) [3]
- PROMIS-Anxiety T-Score 40.9 (Mild) – 85.2 (Severe)
- PROMIS-Depression 41.0 (Mild) – 79.4 (Severe)

Results

- 447 HS patients (mean age 36 years, 79.4% female; 51.8% Non-Hispanic White among US patients) were included.
- HSSA Question 8 response quartiles:

0	1-2	3-6	7-10
Quartile 1, N=125	Quartile 2, N=103	Quartile 3, N=109	Quartile 4, N=110

Figure 1 Odor severity distribution by sex

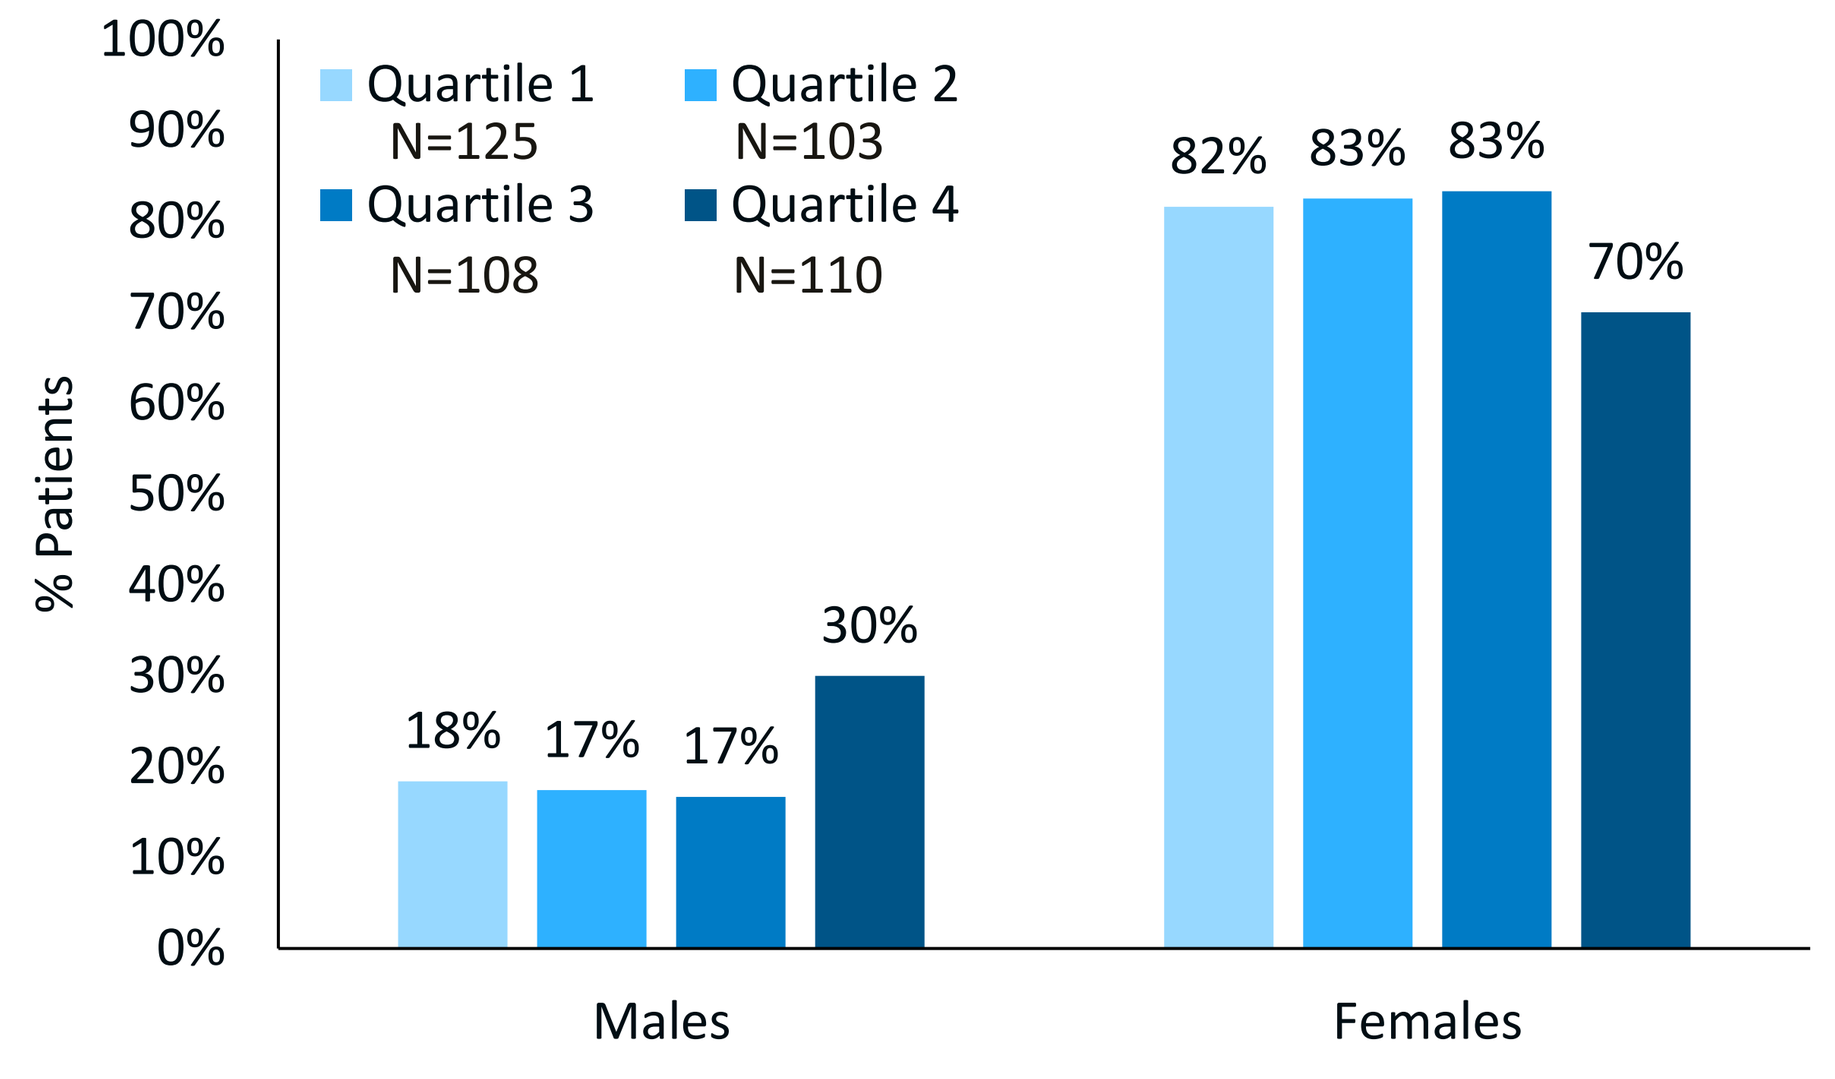
Quartile 1 N=125
Quartile 2 N=103
Quartile 3 N=108
Quartile 4 N=110
% Patients
18% 17% 17% 30%
82% 83% 83% 70%
Males
Females

Figure 2 Mean disease duration (years) by odor severity

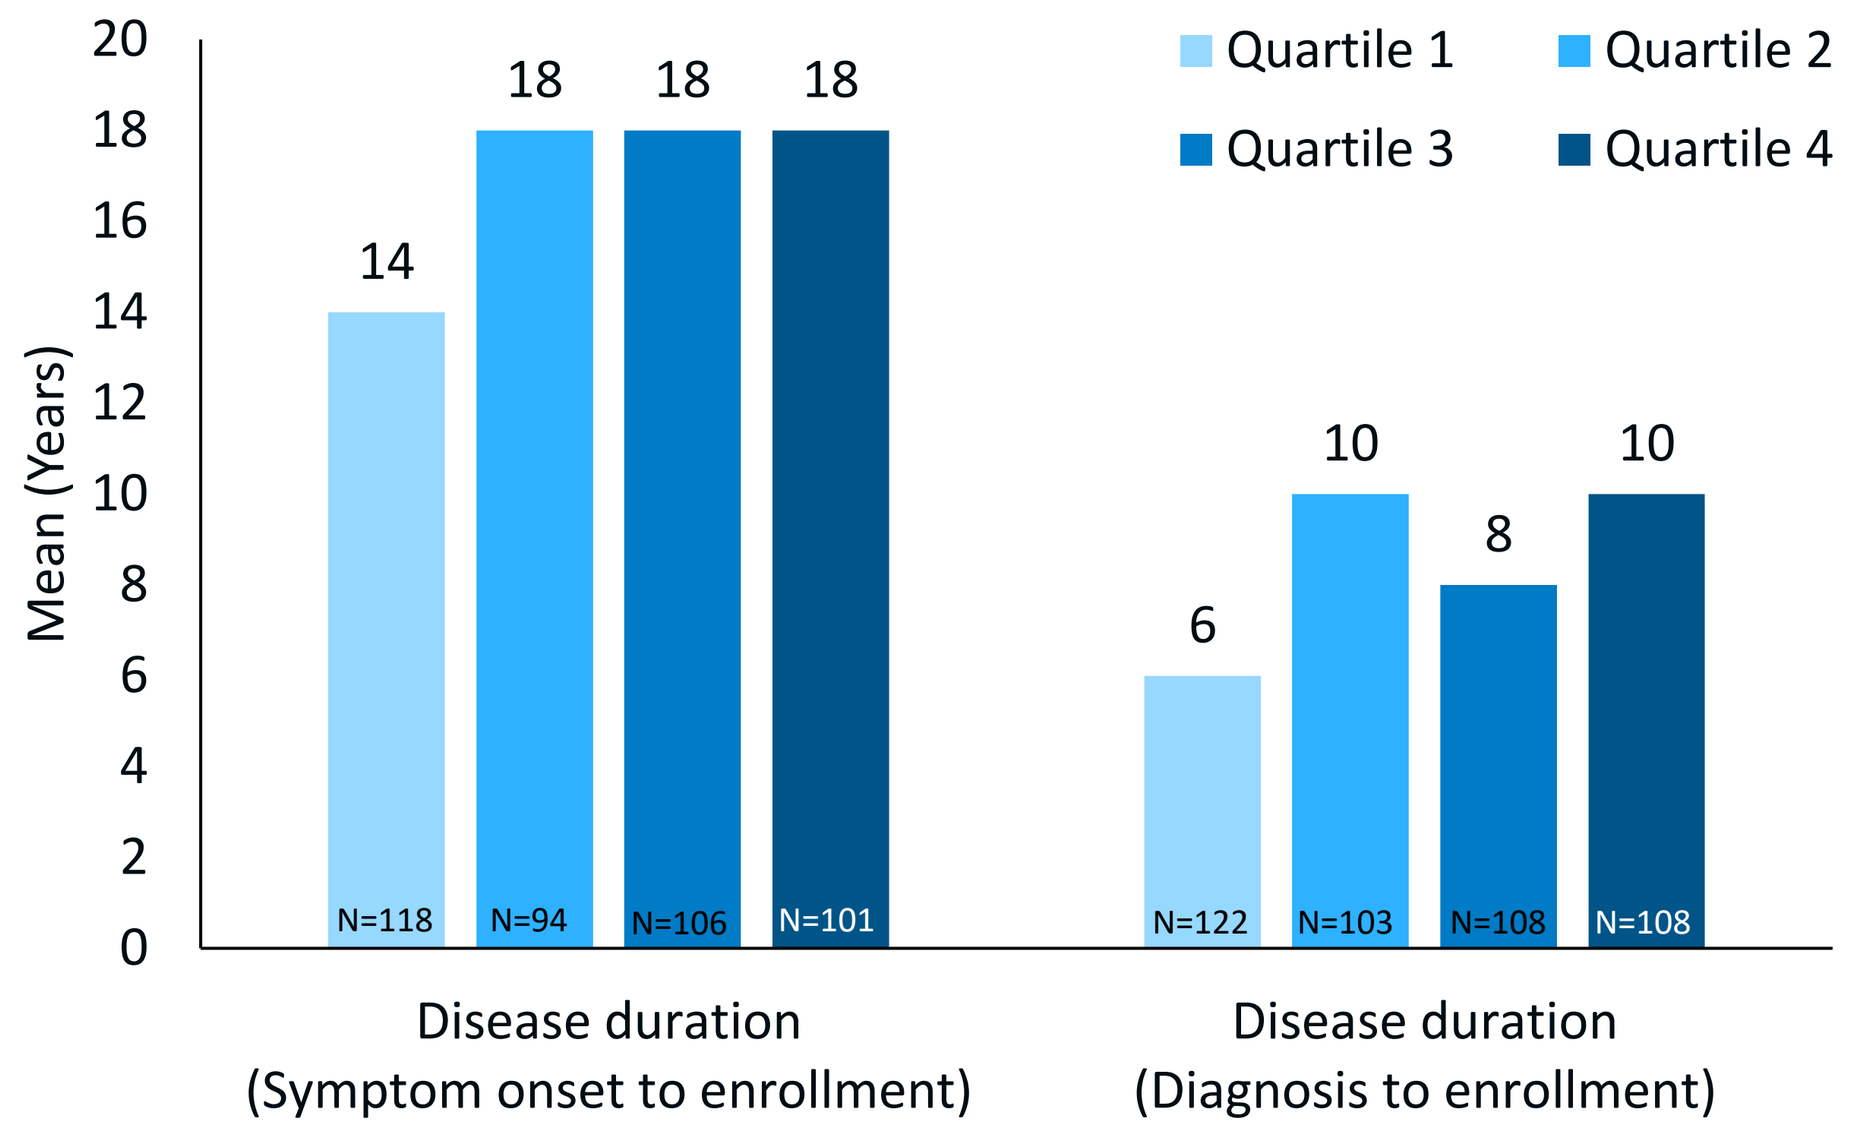
Quartile 1
Quartile 2
Quartile 3
Quartile 4
Mean (Years)
14 18 18 18
N=118 N=94 N=106 N=101
6 10 8 10
N=122 N=103 N=108 N=108
Disease duration (Symptom onset to enrollment)
Disease duration (Diagnosis to enrollment)

- Among males and patients with longer disease duration, a higher proportion were more likely to report increased odor severity (p<.05, Figures 1 and 2).
- Odor severity did not vary by age group or obesity status (not shown).

Figure 3 Odor severity distribution by Hurley stage

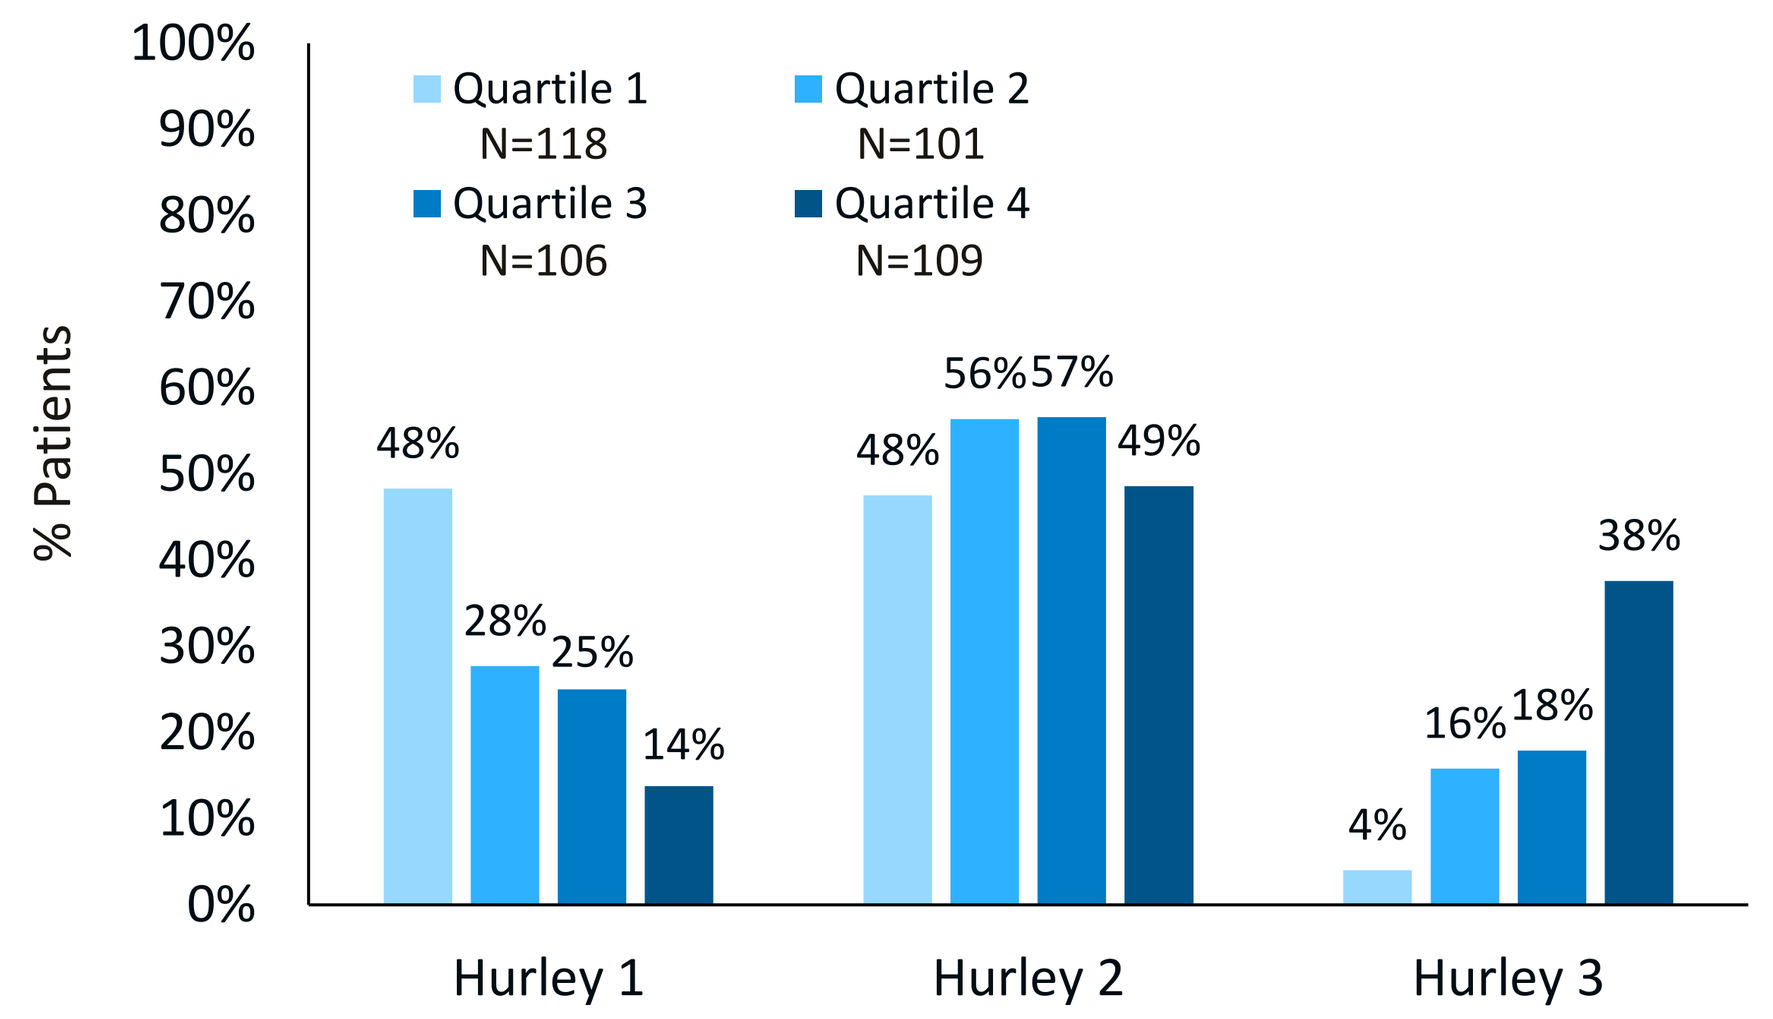
Quartile 1 N=118
Quartile 2 N=101
Quartile 3 N=106
Quartile 4 N=109
% Patients
48% 28% 25% 14%
48% 56% 57% 49%
4% 16% 18% 38%
Hurley 1
Hurley 2
Hurley 3

Figure 4 HS-PGA distribution by odor severity subgroup

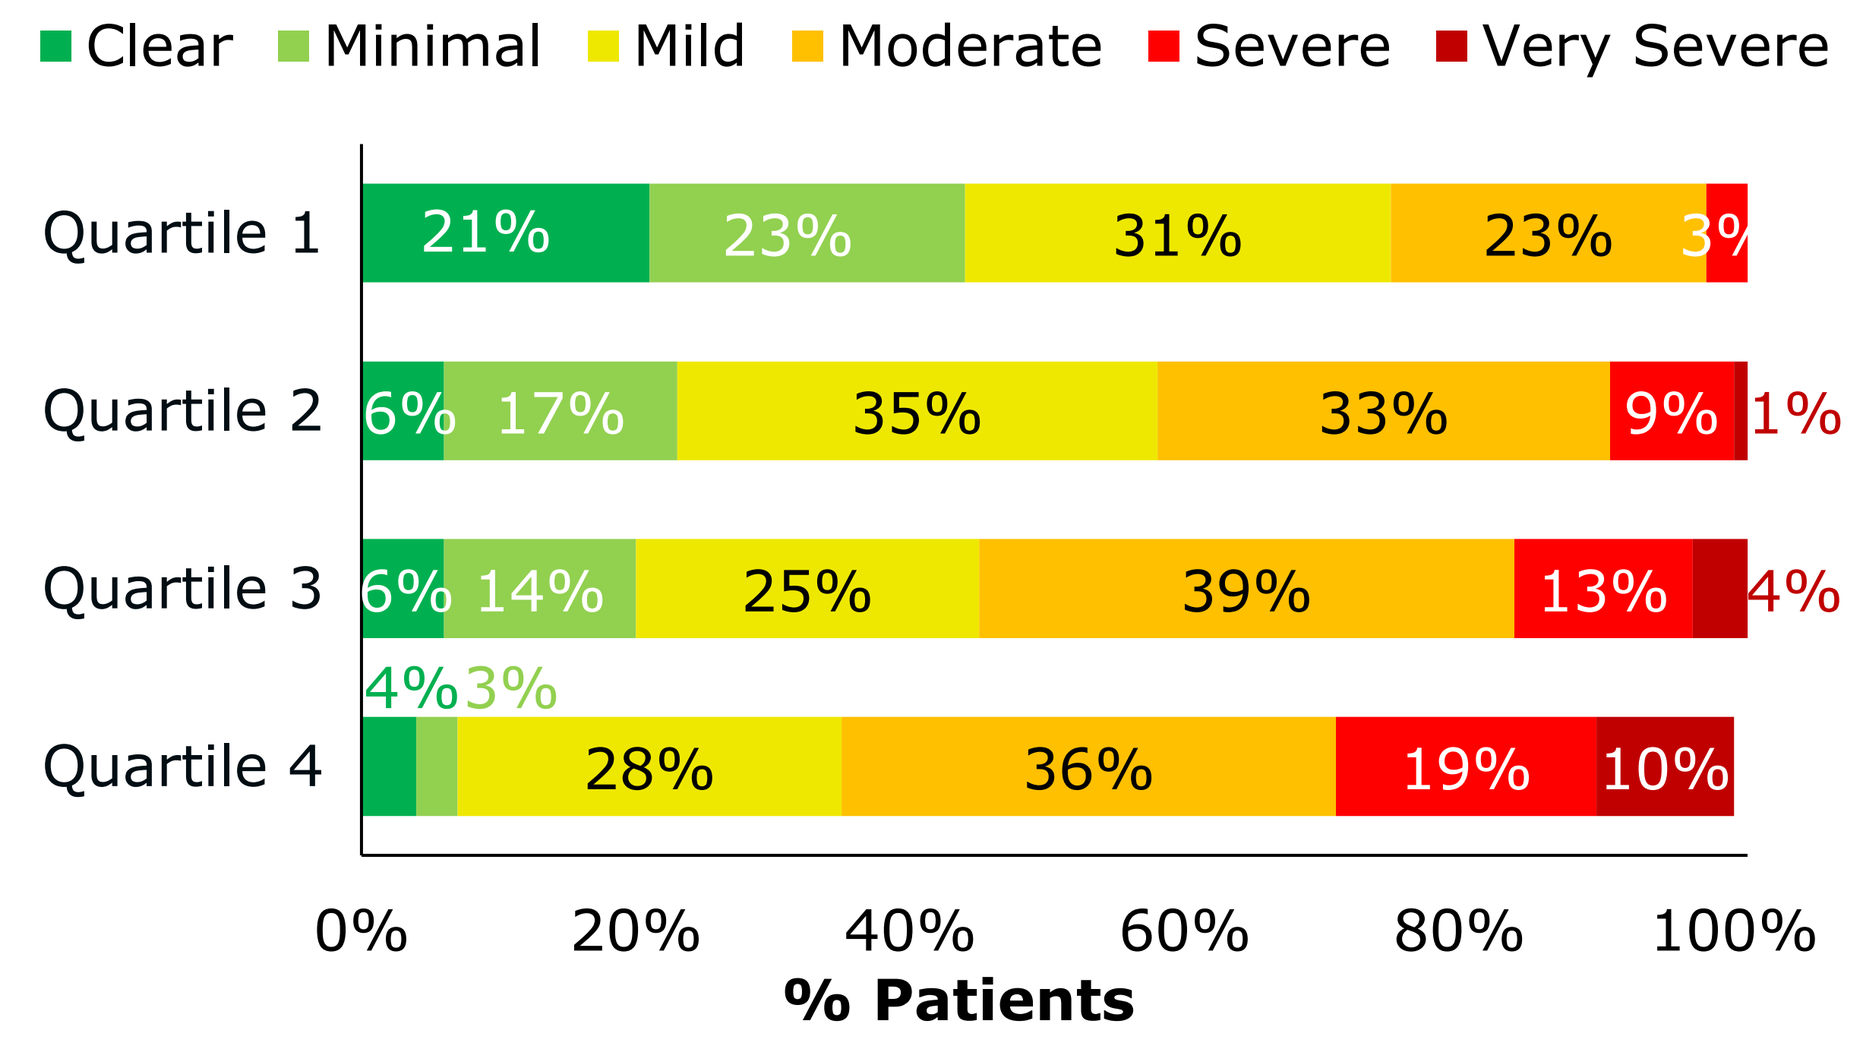
Clear
Minimal
Mild
Moderate
Severe
Very Severe
Quartile 1 21% 23% 31% 23% 3%
Quartile 2 6% 17% 35% 33% 9% 1%
Quartile 3 6% 14% 25% 39% 13% 4%
Quartile 4 4% 3% 28% 36% 19% 10%
% Patients

- Trend testing showed that as the disease severity increased, odor severity was significantly higher, based on Hurley stage and HS-PGA (p<.0001, Figures 3 & 4), and the number of HS anatomical locations (p<.002, Figure 5).
- Odor severity increased with an increasing number of HS lesions (p<.0001).
- Surprisingly, even in patients without abscesses or fistulas, odor severity increased with a higher nodule count (Figure 6).
- Patients currently experiencing a flare and those reporting more severe drainage had greater odor severity (Figure 7 and p<.0001, Figure 8).
- Higher total scores on the HSSA and HiSQoL were associated with worse odor severity (p<0.0001), highlighting the need to explore the impact of odor on quality of life.
- After adjusting for covariates, the models confirmed that odor severity significantly affects HiSQoL, DLQI, and PROMIS Depression and Anxiety scores.

Figure 5 Mean number of body sites with HS involvement by odor severity subgroup

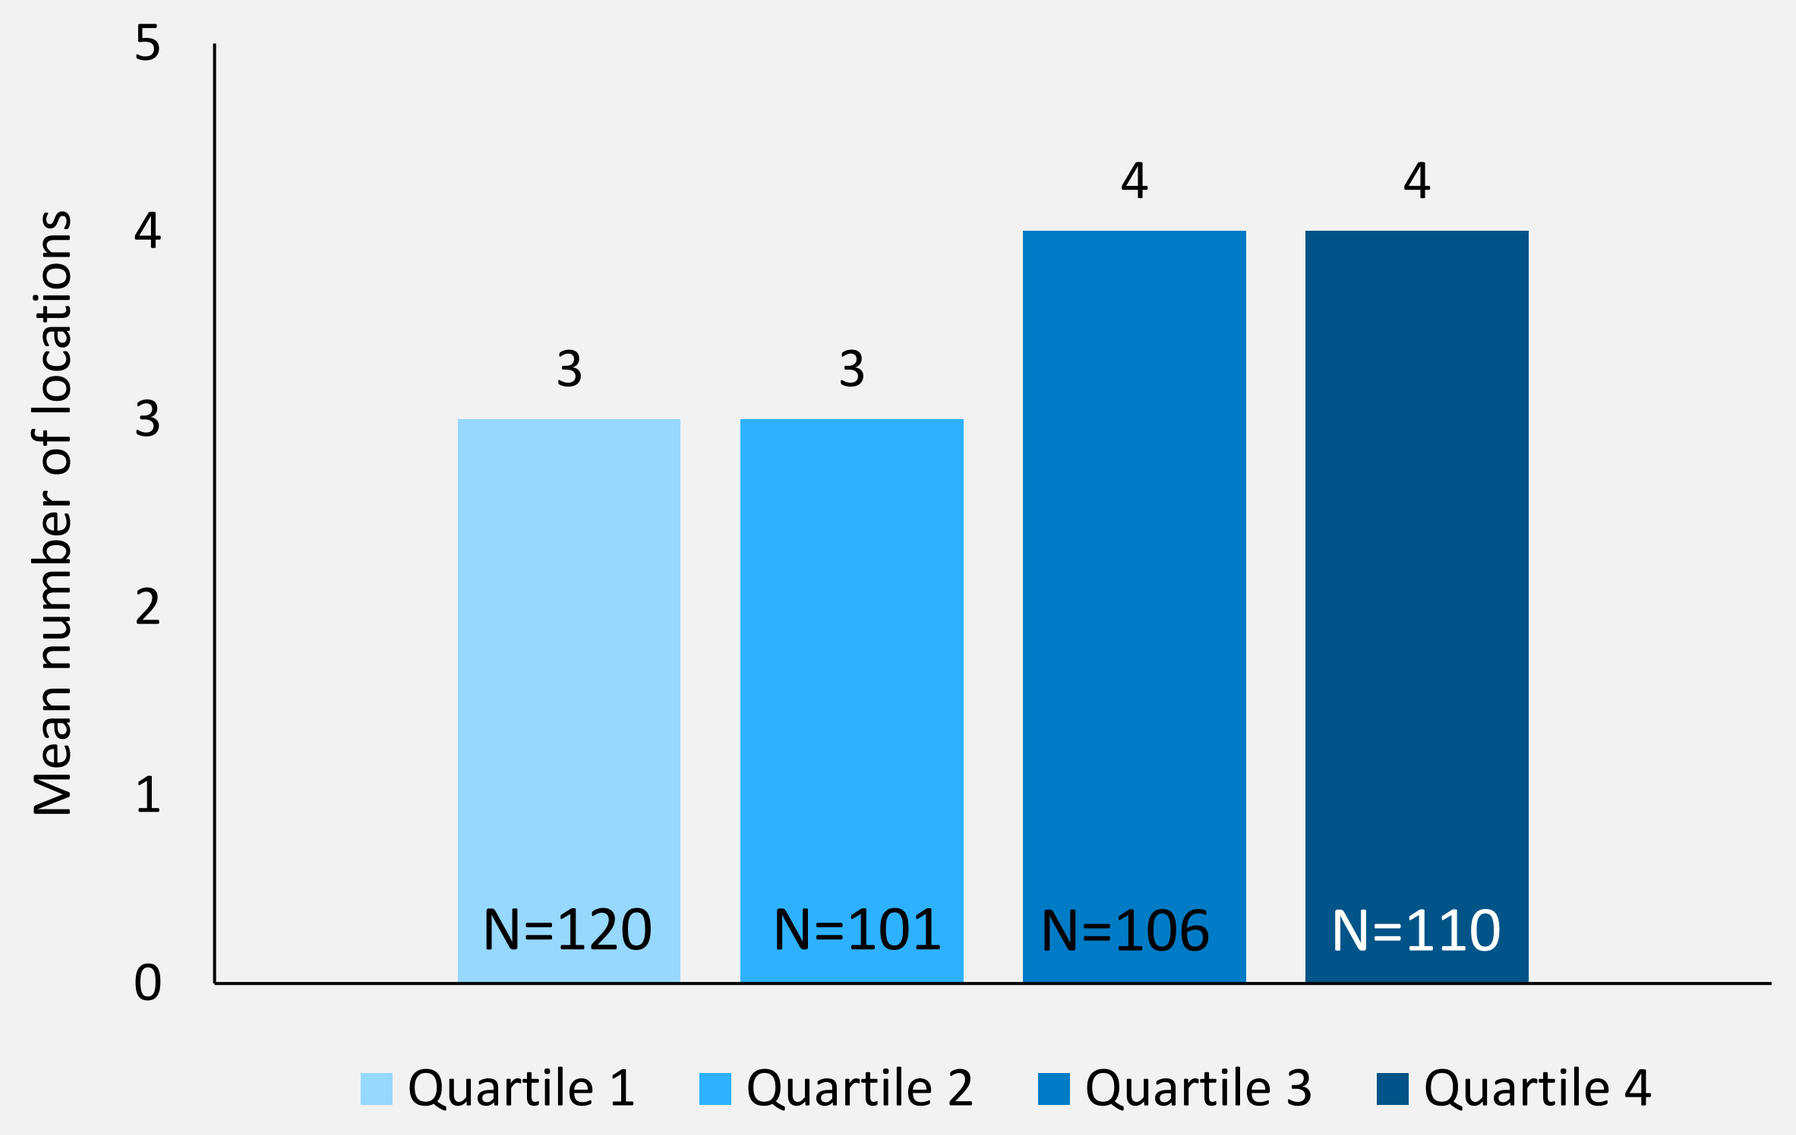
Mean number of locations
3 3 4 4
N=120 N=101 N=106 N=110
Quartile 1
Quartile 2
Quartile 3
Quartile 4

Figure 6 Mean lesion count by type and odor severity subgroup

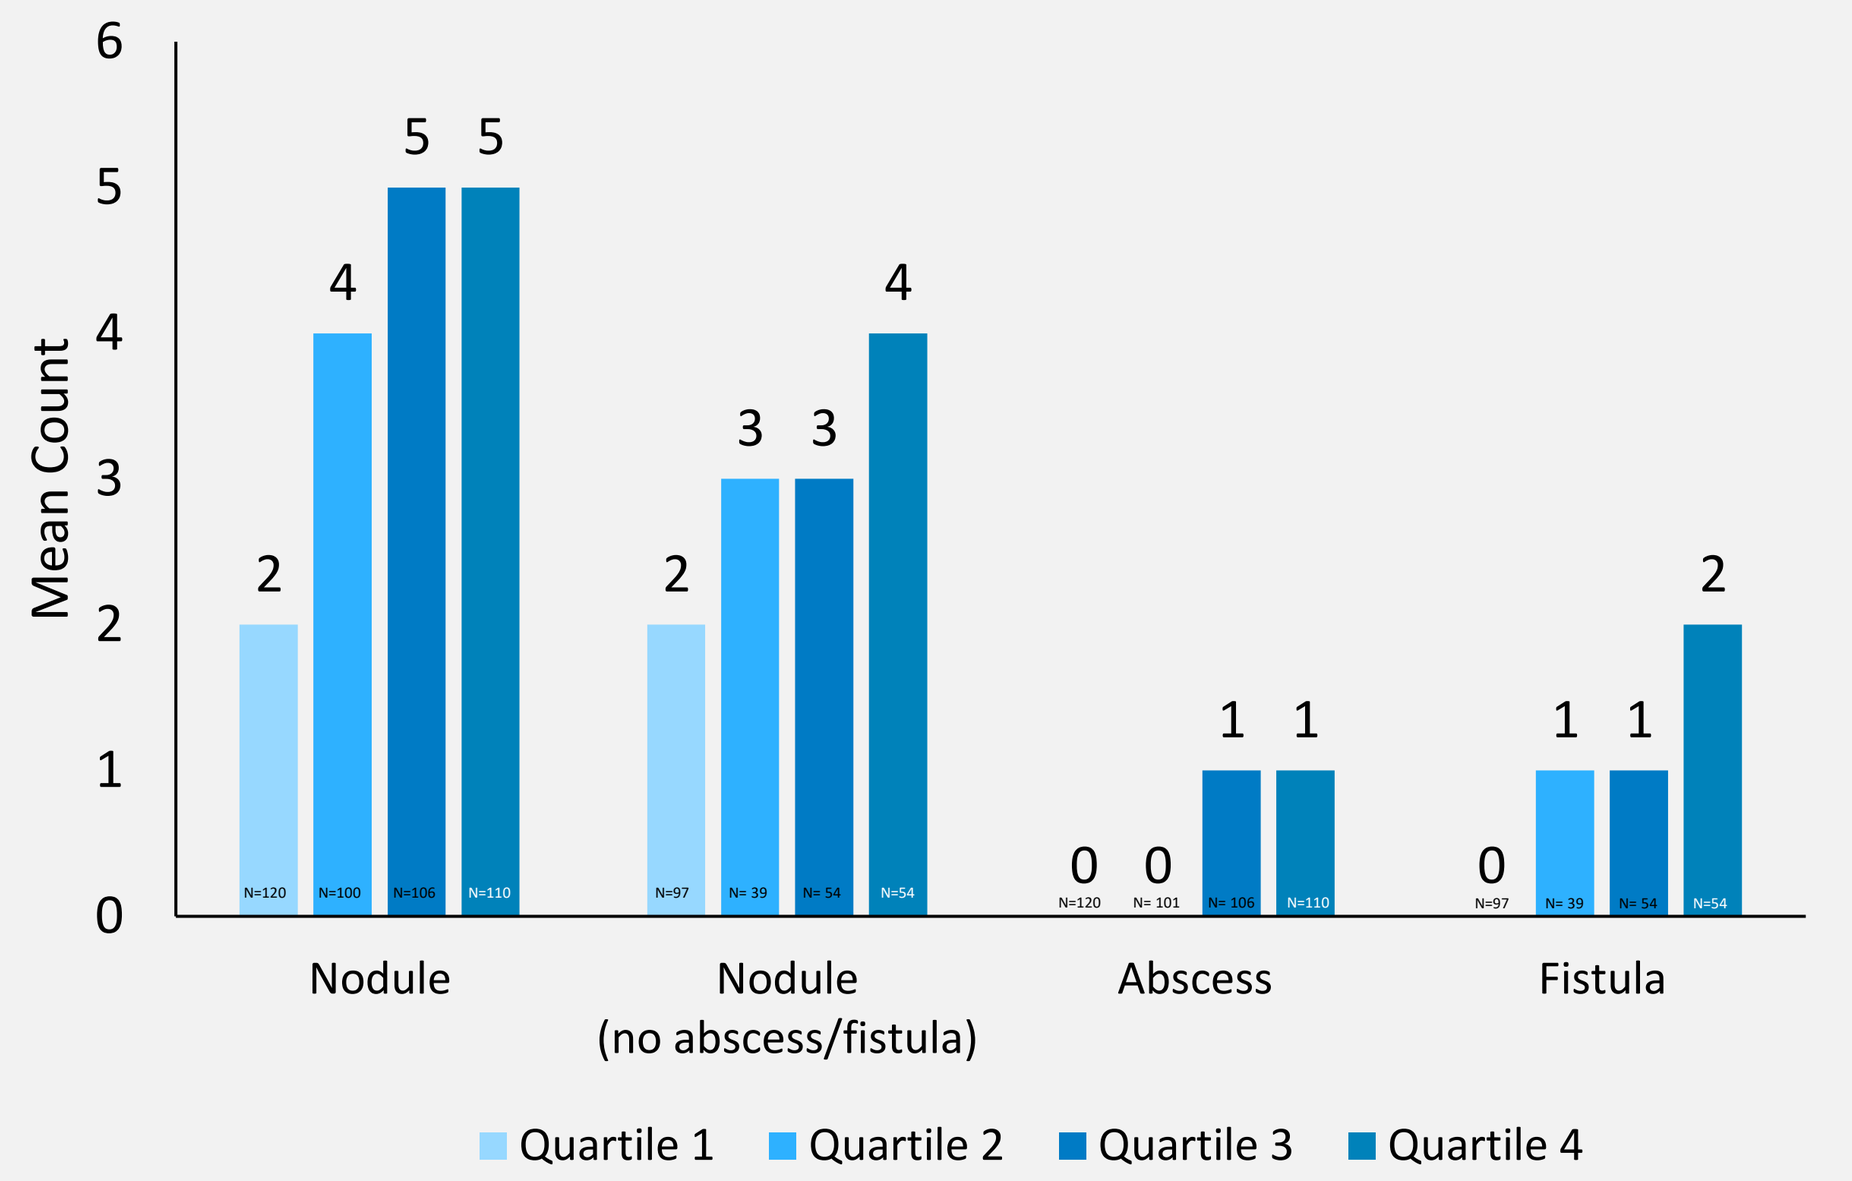
Mean Count
2 4 5 5
2 3 3 4
0 0 1 1
0 1 1 2
Nodule
Nodule (no abscess/fistula)
Abscess
Fistula
Quartile 1
Quartile 2
Quartile 3
Quartile 4

Figure 7 Percentage of patients currently flaring by odor severity subgroup

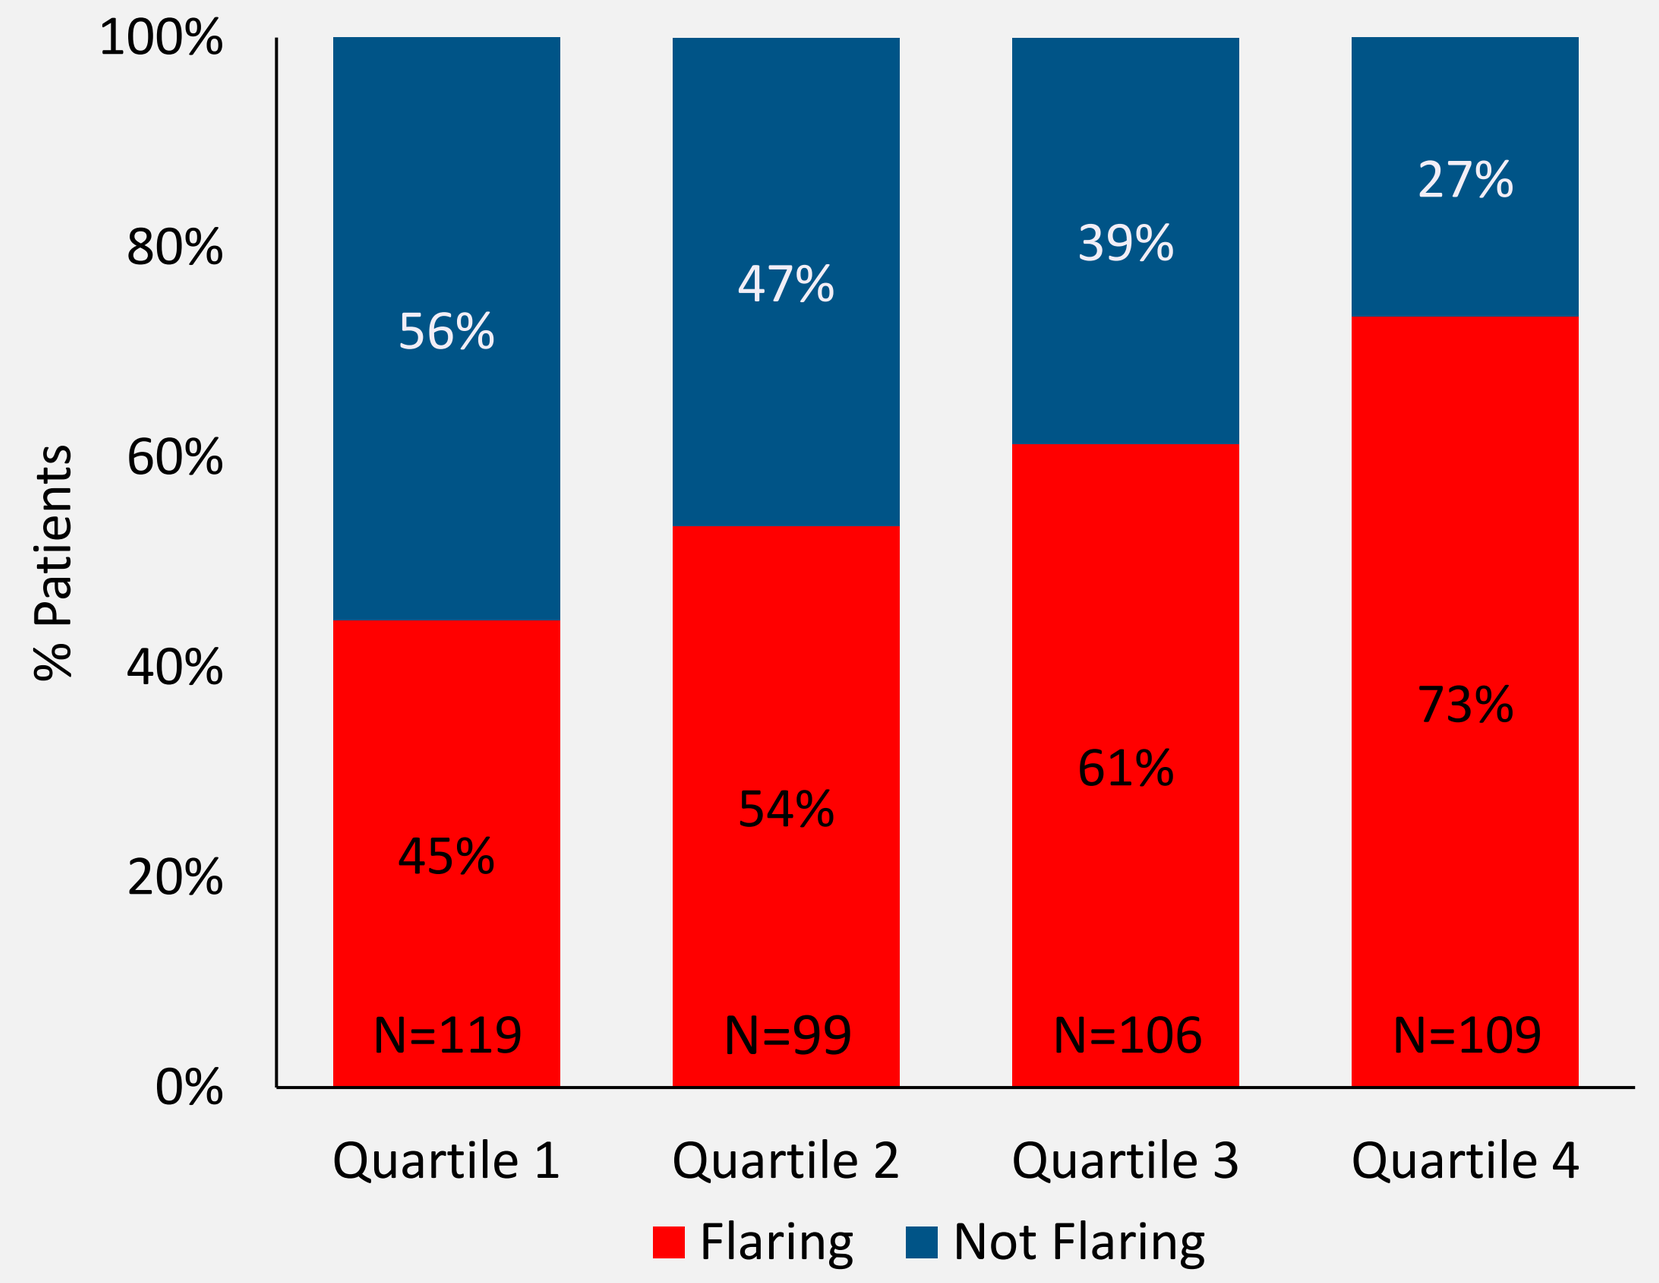
% Patients
56% 47% 39% 27%
45% 54% 61% 73%
N=119 N=99 N=106 N=109
Quartile 1
Quartile 2
Quartile 3
Quartile 4
Flaring
Not Flaring

Figure 8 Mean worst drainage score by odor severity subgroup

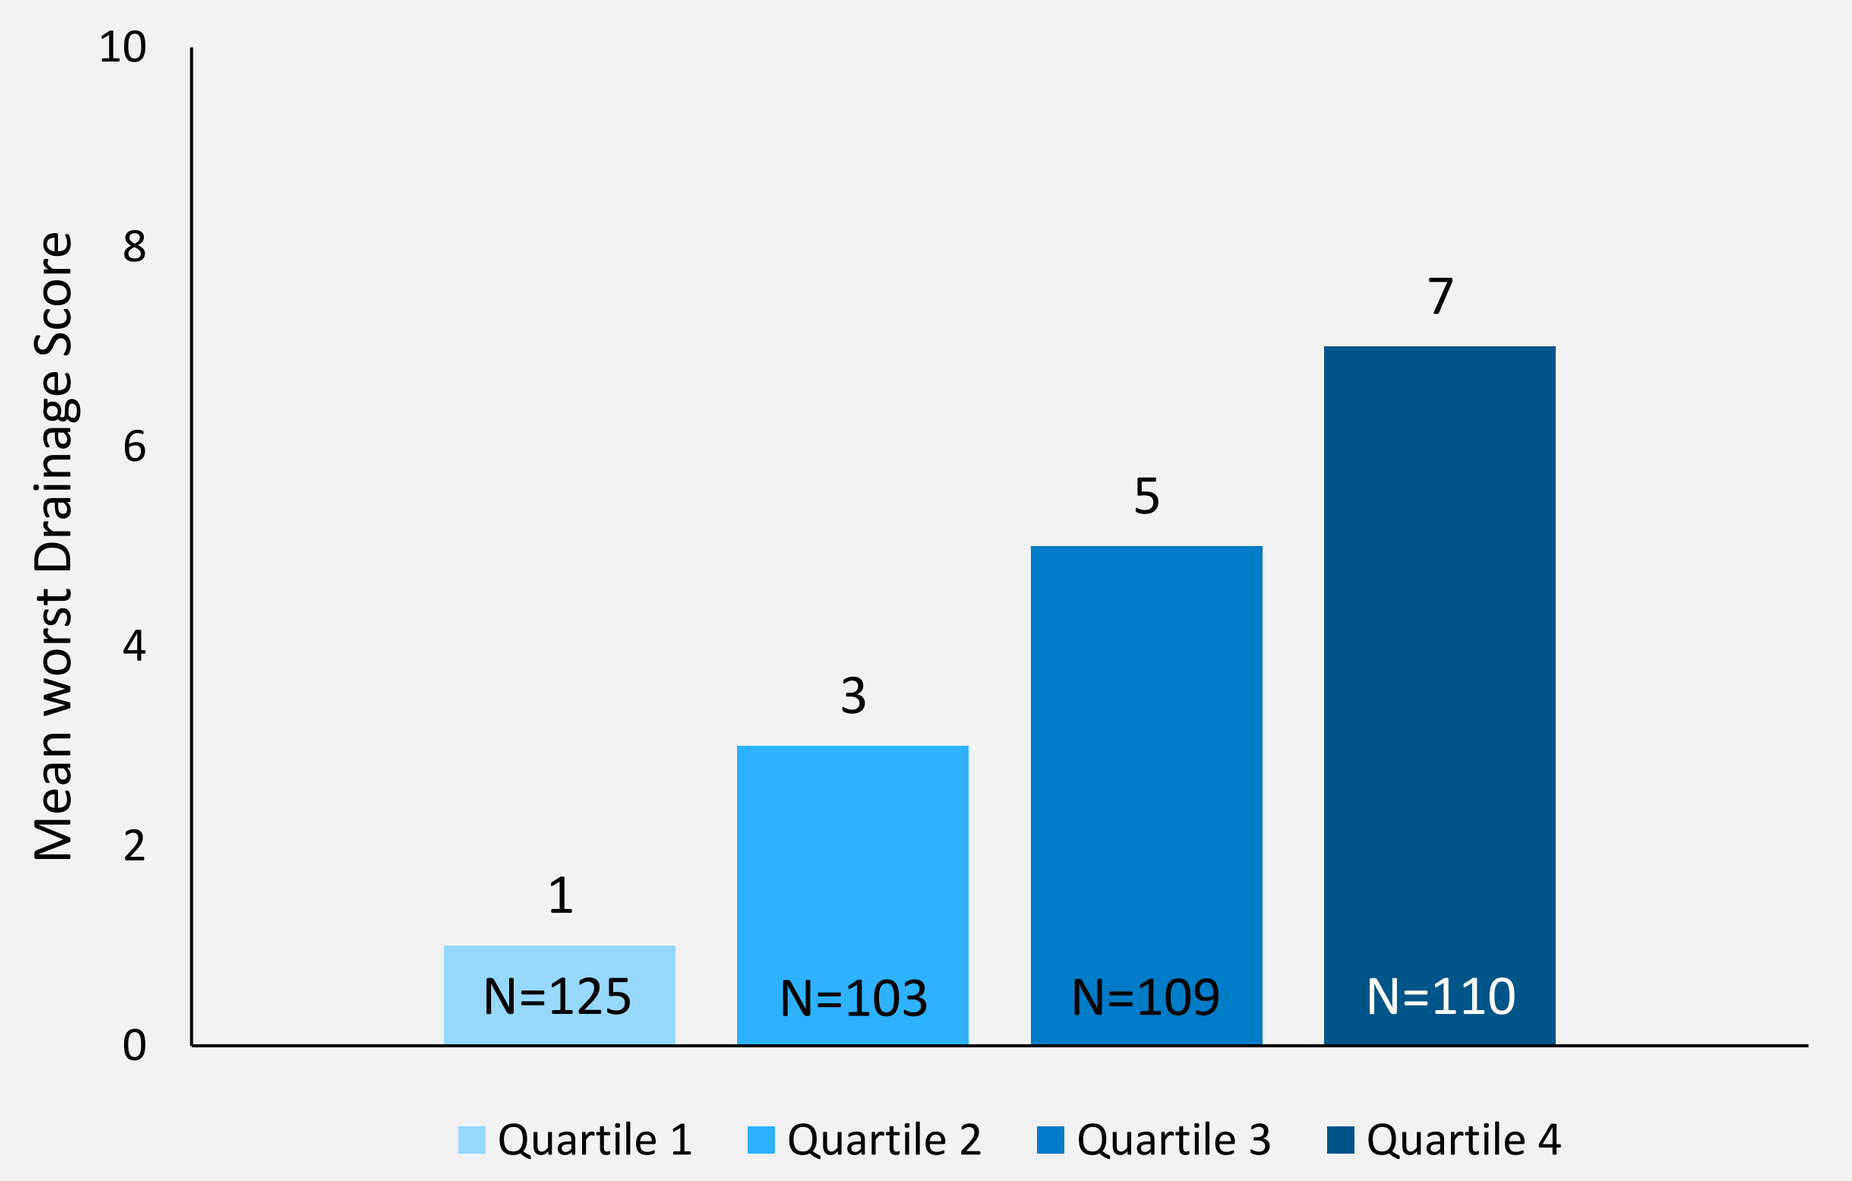
Mean worst Drainage Score
1 3 5 7
N=125 N=103 N=109 N=110
Quartile 1
Quartile 2
Quartile 3
Quartile 4

Conclusions

Given the frequency and severity of odor reported by HS patients across multiple demographics and disease characteristics, these data highlight the importance of assessing odor severity and its impact on patients' QoL.

References

1. Kimball, A.B., et al., *Development and initial psychometric evaluation of patient-reported outcome questionnaires to evaluate the symptoms and impact of hidradenitis suppurativa.* J Dermatolog Treat, 2018. **29**(2): p. 152-164.
2. Kimball, A.B., et al. *Real-world study of hidradenitis suppurativa: skin lesions and pain are associated with Hurley Stage in TARGET-DERM HS*. in *Maui Derm*. 2024. Maui, HI.
3. Kirby, J.S., et al., *The Hidradenitis Suppurativa Quality of Life (HiSQoL) score: development and validation of a measure for clinical trials.* Br J Dermatol, 2020. **183**(2): p. 340-348.

Acknowledgements and Disclosures: Disclosure of Industry Support: Target RWE communities are collaborations among academic & community investigators, the pharmaceutical industry, and patient community advocates. Target RWE communities are sponsored by TARGET PharmaSolutions Inc (d.b.a., Target RWE). The authors would like to thank all the investigators, participants, and research staff associated with TARGET-DERM HS. ClinicalTrials.gov Identifier: NCT03661866.AbbVie Inc. subscribed to TARGET-DERM HS.
TM has received clinical research and grants to university from: Acelyrin, BMS, ChemoCentryx, Galderma, Janssen, Lilly, Pfizer, Boehringer Ingelheim, Incyte, Regeneron, and Moonlake. She has consulted for: Abbvie, Arcutis, BMS, Bodewell, Janssen, Lilly, Leo Novartis, Pfizer, Physician Education, Resources (PER), UCB. **JP, BS, BR, AG, CB, and AD** are employees of AbbVie Inc. and may hold stock options. **FY** is an employee of Target RWE and may hold stock options. **BHK** has been an investigator for Biogen, InflaRx, NIAMS, Merck, and Janssen. He was also a consultant for: Eli Lilly, Novartis, Biogen, and ADC Therapeutics. **MC** is an investigator for Target RWE. **AK** her institution has received grants from AbbVie, Admirx, AnaptysBio, Aristea, Bristol Myers Squibb, Eli Lilly, Incyte, Janssen, MoonLake, Novartis, Pfizer, Prometheus, UCB, and Sonoma Bio, fellowship funding from Janssen and AbbVie. She received consulting fees from AbbVie, Alumis, Bayer, Boehringer Ingelheim, Eli Lilly, Evommune, Janssen, MoonLake, Novartis, Pfizer, Priovant, Sonoma Bio, Sanofi, UCB, and Target RWE, and serves on the board of directors of Almirall.

American Academy of Dermatology • March 7-11, 2025 • Orlando, FL