AASLD 2024 TARGET MASH poster v2.3 B OUTLINE MECHANICAL.pdf 1 10/25/24 11:34 AM

Understanding Metabolic Dysfunction-Associated Steatohepatitis: Differences in Clinical and Demographic Characteristics Across Racial and Ethnic Groups

Andrea R. Mospan, PhD, RAC[1]; Breda Munoz, PhD[1]; Heather L. Morris, PhD[1]; Aarth Sheth, PharmD[2]; Rakesh Luthra, MS[2] on behalf of the TARGET-NASH investigators

[1]Target RWE, Durham, NC; [2]Novo Nordisk Inc., Plainsboro, NJ.

https://sciencehub.novonordisk.com/aasld2024/Sheth.html?cid=qr-xi8e47ui12

Plain Language Summary

- **Why it matters?** Metabolic dysfunction-associated steatohepatitis (MASH) is a serious liver disease that can lead to progressive fibrosis, cirrhosis, and other complications. Among those with MASH in the United States, studies have shown health disparities, or differences in health and health care experienced by socially and economically disadvantaged racial and ethnic groups, exist
- **How it works?** Data from the TARGET-NASH cohort were analyzed to describe the differences in health and health care between racial and ethnic groups among people with MASH in the United States
- **What we found?** Among people with MASH in the United States, significant differences in demographics, measures of health, and health care were observed between different racial and ethnic groups

Introduction

- MASH is estimated to affect 1.5% to 6.5% of US adults[1]
- In the US, studies have shown significant racial disparities in the prevalence and severity of MASH[2]
- This analysis assessed health disparities across racial and ethnic groups among patients with non-cirrhotic MASH and MASH cirrhosis (referred to as "cirrhosis" throughout the poster) enrolled in TARGET-NASH

Methods

- Adults (aged ≥18 years) with non-cirrhotic MASH or cirrhosis enrolled in TARGET-NASH were assessed for demographic/clinical characteristics, diagnostic testing, and all-cause mortality by racial and ethnic group (Hispanic/Latino, non-Hispanic White, non-Hispanic Black, and non-Hispanic Other/not reported)
 - TARGET-NASH, sponsored by Target RWE, is an ongoing longitudinal real-world cohort of >6500 patients with metabolic dysfunction-associated steatotic liver disease (MASLD) who are managed at academic and community sites in the United States[3]
- The index date was defined as the earliest date evidence of MASH was observed during the 3-year retrospective period prior to enrollment, and the study period was August 2016 to November 2023
- Outcomes were examined descriptively at index. Incidence rates (IRs) and proportions were determined for post-index all-cause mortality

Results

- The cohort in this analysis included 2738 adults with non-cirrhotic MASH or cirrhosis and was comprised of 325 (11.9%) Hispanic/Latino patients, 1999 (73.0%) non-Hispanic White patients, 145 (5.3%) non-Hispanic Black patients, and 269 (9.8%) non-Hispanic Other patients (**Table 1**)

Results (cont'd)

- Insurance type was significantly different across racial and ethnic groups with highest Medicaid use among non-Hispanic Black patients (*P*<.01)
- Rates of overweight (BMI >25 kg/m^2) and obesity (BMI >30 kg/m^2) were highest among non-Hispanic Black patients

Table 1: Demographics at index stratified by racial and ethnic groups among patients with non-cirrhotic MASH or cirrhosis enrolled in TARGET-NASH

	Hispanic/ Latino n=325	Non-Hispanic White n=1999	Non-Hispanic Black n=145	Non-Hispanic Other[a] n=269	*P*[b]	All N=2738
Age, years						
Mean (SD)	53.5 (14.0)	57.5 (12.7)	54.3 (11.9)	55.5 (13.8)	<.01	56.7 (13.0)
Categorical age, n (%)					<.01	
18–64 years	248 (76.3)	1336 (66.8)	117 (80.7)	197 (73.2)		1898 (69.3
≥65 years	77 (23.7)	663 (33.2)	28 (19.3)	72 (26.8)		840 (30.7)
Sex, n (%)					<.01	
Male	122 (37.5)	843 (42.4)	42 (29.0)	145 (53.9)		1152 (42.1)
Female	203 (62.5)	1156 (57.8)	103 (71.0)	124 (46.1)		1586 (57.9)
Insurance type, n (%)					<.01	
Medicaid	31 (9.5)	89 (4.5)	16 (11.0)	20 (7.4)		156 (5.7)
Medicare	58 (17.8)	433 (21.7)	24 (16.6)	46 (17.1)		561 (20.5)
Other	2 (0.6)	10 (0.5)	0 (0.0)	3 (1.1)		15 (0.5)
Private	144 (44.3)	932 (46.6)	63 (44.1)	131 (48.7)		1271 (46.4)
Supplemental	0 (0.0)	4 (0.2)	0 (0.0)	1 (0.4)		5 (0.2)
Uninsured	7 (2.2)	15 (0.8)	2 (1.4)	1 (0.4)		25 (0.9)
Unknown	83 (25.5)	516 (25.8)	39 (26.9)	67 (24.9)		705 (25.7)
BMI, kg/m^2						
n	322	1976	144	268		2710
Mean (SD)	33.1 (6.44)	34.4 (7.46)	36.2 (8.66)	29.8 (6.85)	<.01	33.9 (7.51)
Categorical BMI, n (%)					<.01	
≤25, kg/m^2	22 (6.8)	124 (6.3)	8 (5.6)	61 (22.8)		215 (7.9)
>25-30, kg/m^2	95 (29.5)	461 (23.3)	28 (19.4)	102 (38.1)		686 (25.3)
>30-35, kg/m^2	105 (32.6)	576 (29.1)	34 (23.6)	58 (21.6)		773 (28.5)
>35-40, kg/m^2	54 (16.8)	415 (21.0)	37 (25.7)	28 (10.4)		534 (19.7)
>40, kg/m^2	46 (14.3)	400 (20.2)	37 (25.7)	19 (7.1)		502 (18.5)
Follow-up time, months						
Mean (SD)	60.4 (20.23)	60.5 (21.64)	64.2 (21.13)	61.3 (20.62)	0.19	60.8 (21.36)

BMI, body mass index; MASH, metabolic dysfunction-associated steatohepatitis; NASH, nonalcoholic steatohepatitis.

[a]Non-Hispanic Other includes patients without reported racial and ethnic group data; [b]Chi-square or Fisher exact test comparing categorical variables or Kruskal-Wallis comparing distribution of continuous variables.

- Rates of hypertension, type 2 diabetes, and hyperlipidemia were highest among non-Hispanic Black patients, while rates of cardiovascular (CV) disease/events were highest among non-Hispanic White patients (**Figure 1**)
- Ultrasound and vibration-controlled transient elastography (FibroScan®) were the most prevalent diagnostic testing methods used by >45% of all patients

Figure 1: Comorbidities present at index stratified by racial and ethnic group among patients with non-cirrhotic MASH or cirrhosis enrolled in TARGET-NASH

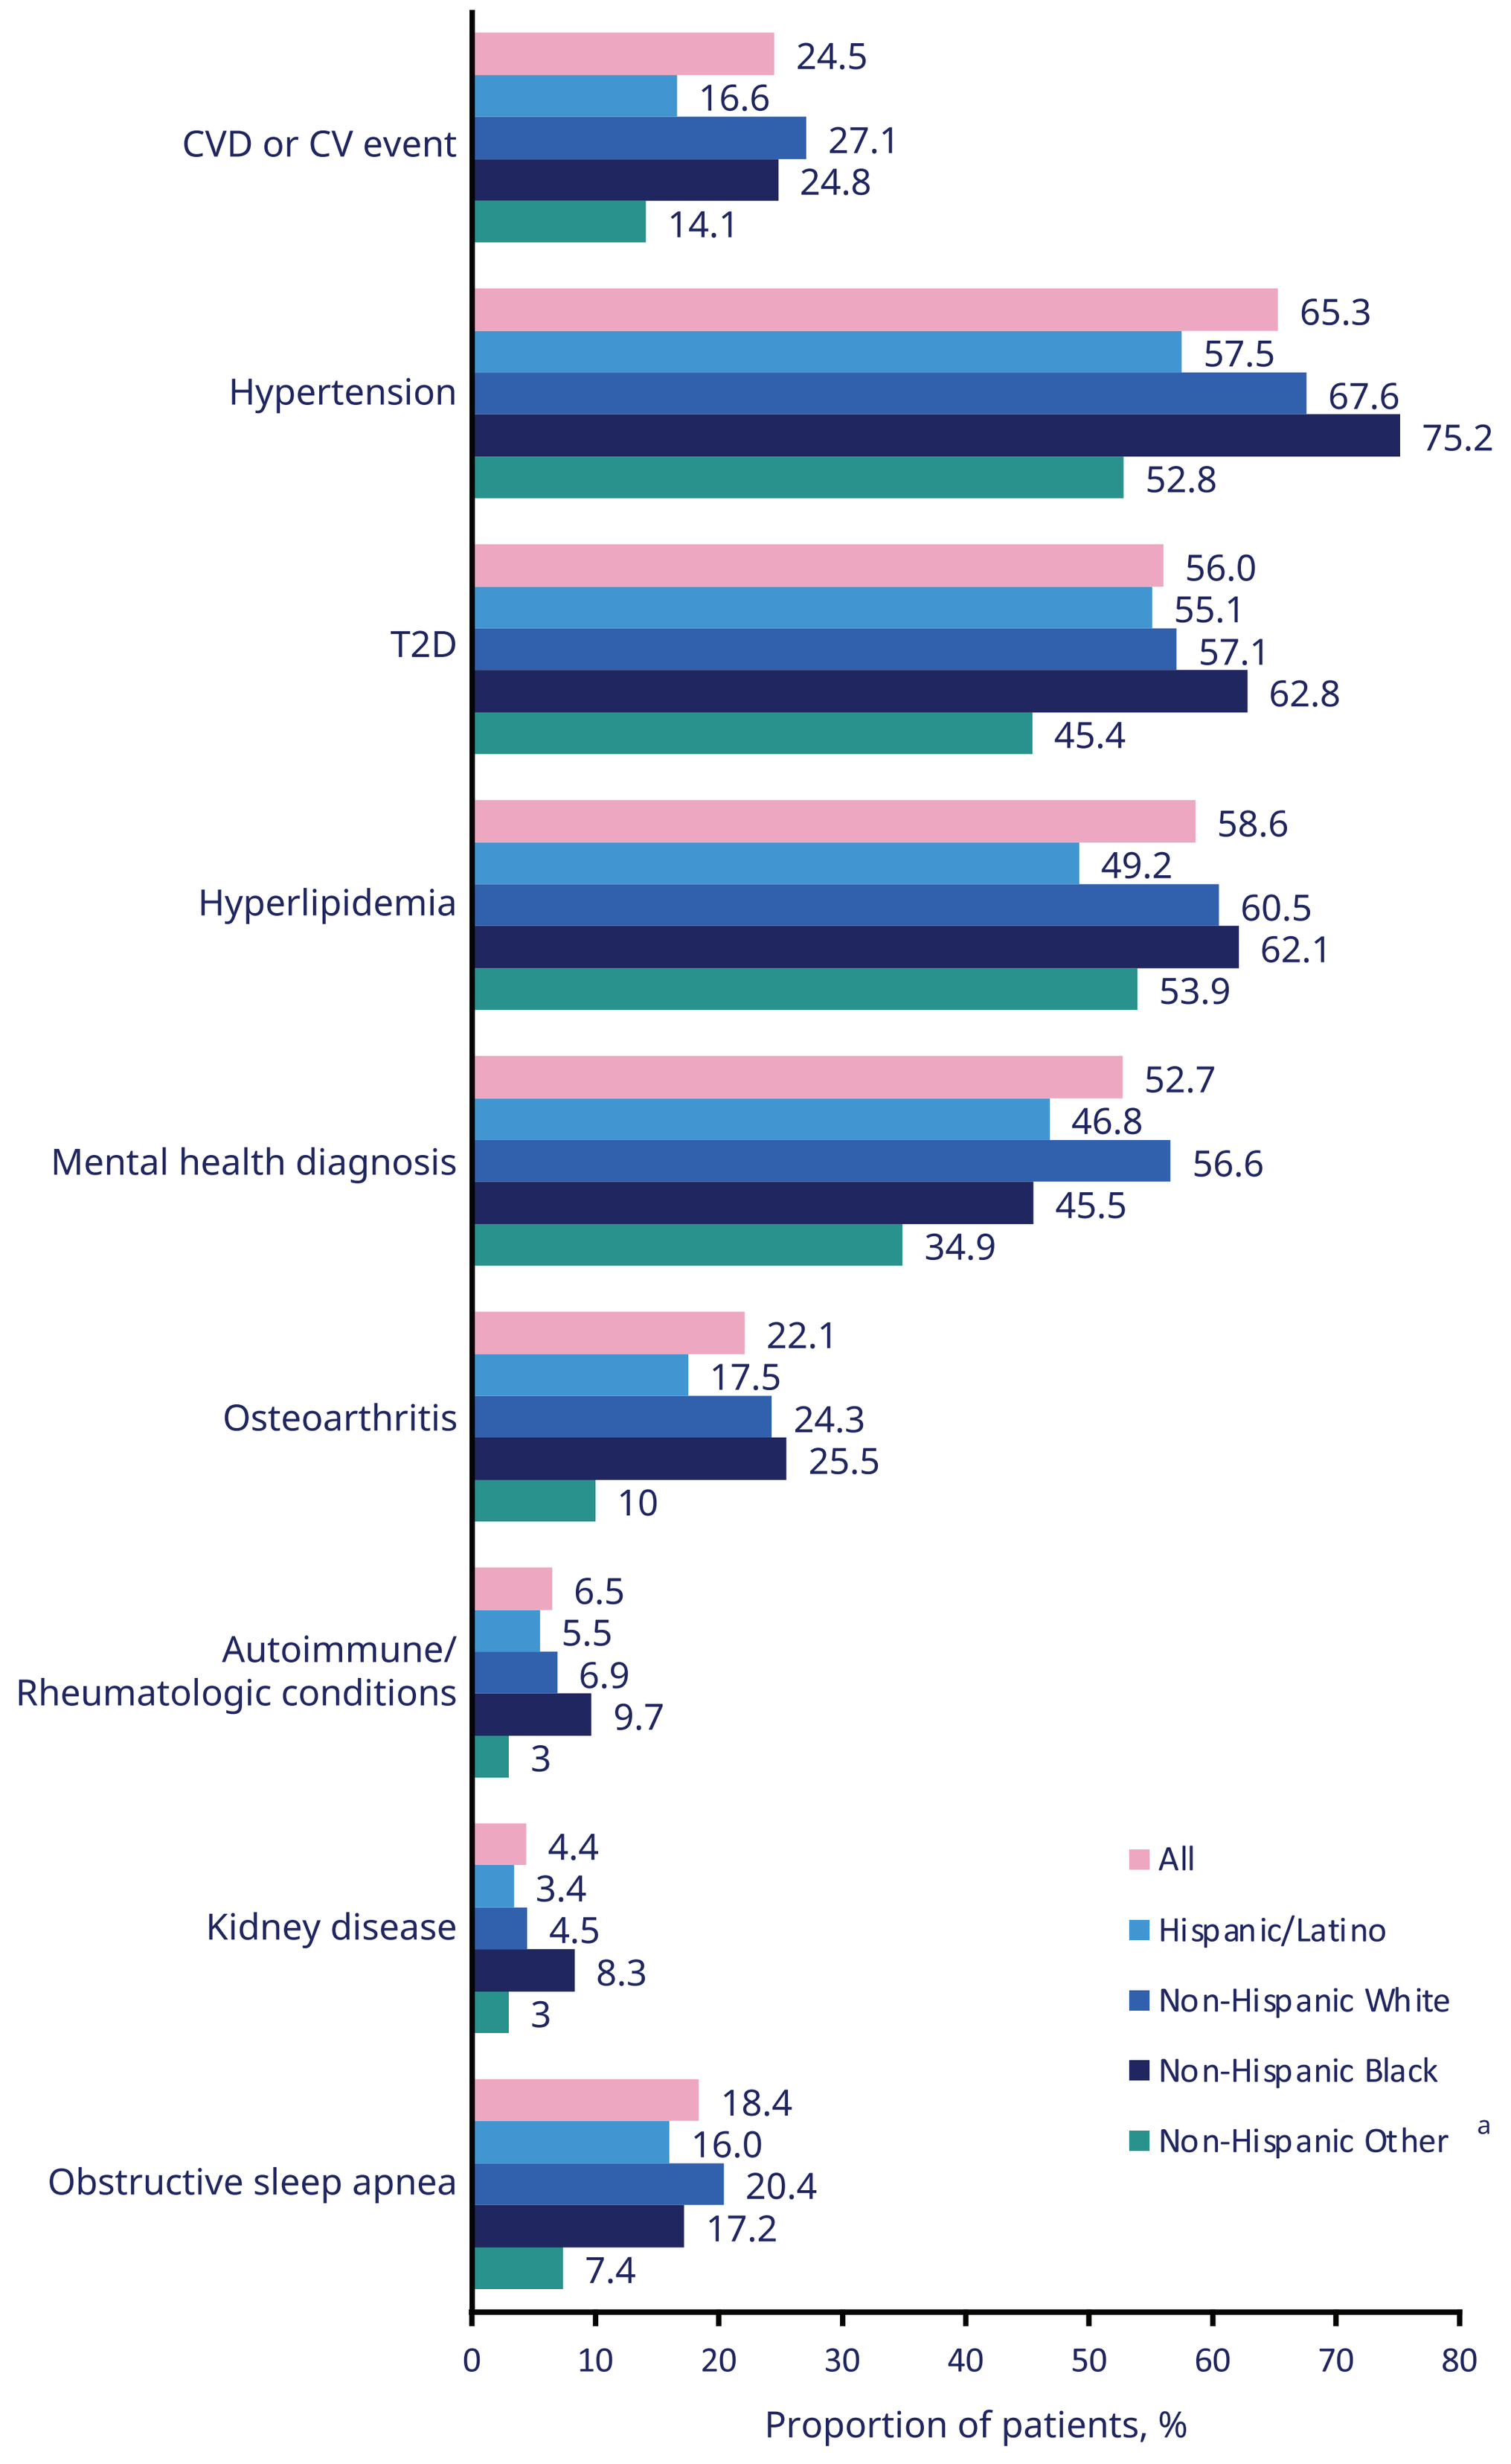
CVD or CV event
24.5
16.6
27.1
24.8
14.1
Hypertension
65.3
57.5
67.6
75.2
52.8
T2D
56.0
55.1
57.1
62.8
45.4
Hyperlipidemia
58.6
49.2
60.5
62.1
53.9
Mental health diagnosis
52.7
46.8
56.6
45.5
34.9
Osteoarthritis
22.1
17.5
24.3
25.5
10
Autoimmune/ Rheumatologic conditions
6.5
5.5
6.9
9.7
3
Kidney disease
4.4
3.4
4.5
8.3
3
Obstructive sleep apnea
18.4
16.0
20.4
17.2
7.4
All
Hispanic/Latino
Non-Hispanic White
Non-Hispanic Black
Non-Hispanic Other a
0 10 20 30 40 50 60 70 80
Proportion of patients, %

CV, cardiovascular; CVD, cardiovascular disease; MASH, metabolic dysfunction-associated steatohepatitis; NASH, nonalcoholic steatohepatitis; T2D, type 2 diabetes.

[a]Non-Hispanic Other includes patients without reported racial and ethnic group data.

- Levels of aspartate aminotransferase (AST) and alkaline phosphatase (AP) were highest among non-Hispanic Black patients (**Table 2**)
- Among patients with biopsy, stage 4 fibrosis was highest in Hispanic/Latino (27.3%) and non-Hispanic White patients (28.1%)
- Medication use differed significantly with highest use of antihypertensives, calcium channel blockers, diuretics, lipid modifying agents, and diabetes medications among non-Hispanic Black patients

Table 2: Laboratory assessments, diagnostic tests, and medication use stratified by racial and ethnic groups among patients with non-cirrhotic MASH or cirrhosis enrolled in TARGET-NASH

	Hispanic/ Latino n=325	Non-Hispanic White n=1999	Non-Hispanic Black n=145	Non-Hispanic Other[a] n=269	*P*[b]	All N=2738
Liver enzyme laboratory assessments at index[c]						
ALT, IU/L					.08	
n	304	1812	141	255		2512
Mean (SD)	68.0 (71.2)	60.0 (59.7)	66.7 (92.9)	67.2 (66.1)		62.1 (64.1)
AST, IU/L					.02	
n	305	1796	139	253		2493
Mean (SD)	52.8 (51.7)	50.7 (43.0)	64.8 (105.4)	49.6 (56.9)		51.6 (51.1)
AP, IU/L					<.01	
n	302	1766	138	251		2457
Mean (SD)	103.8 (53.8)	99.9 (60.6)	111.4 (77.8)	88.2 (52.3)		99.9 (60.3)
Diagnostic test at index, n (%)						
Biopsy	129 (39.7)	757 (37.9)	64 (44.1)	67 (24.9)	<.01	1017 (37.1)
Fibrosis stage[d]					<.01	
n	128	750	62	65		1005
0	36 (28.1)	111 (14.8)	22 (35.5)	15 (23.1)		184 (18.3)
1	26 (20.3)	164 (21.9)	12 (19.4)	18 (27.7)		220 (21.9)
2	11 (8.6)	97 (12.9)	12 (19.4)	11 (16.9)		131 (13.0)
3	20 (15.6)	167 (22.3)	9 (14.5)	11 (16.9)		207 (20.6)
4	35 (27.3)	211 (28.1)	7 (11.3)	10 (15.4)		263 (26.2)
Ultrasound	218 (67.1)	1125 (56.3)	89 (61.4)	211 (78.4)	<.01	1643 (60.0)
CT/MRI	69 (21.2)	417 (20.9)	22 (15.2)	38 (14.1)	<.01	546 (19.9)
VCTE	175 (53.8)	935 (46.8)	81 (55.9)	143 (53.2)	<.01	1334 (48.7)
Medication use at any time during the study, n (%)						
Antihypertensives	40 (12.3)	291 (14.6)	25 (17.2)	20 (7.4)	<.01	376 (13.7)
Beta-blocking agents	109 (33.5)	916 (45.8)	58 (40.0)	88 (32.7)	<.01	1171 (42.8)
Calcium channel blockers	55 (16.9)	480 (24.0)	64 (44.1)	66 (24.5)	<.01	665 (24.3)
Diuretics	125 (38.5)	985 (49.3)	77 (53.1)	66 (24.5)	<.01	1253 (45.8)
Lipid modifying agents	163 (50.2)	1216 (60.8)	92 (63.4)	158 (58.7)	<.01	1629 (59.5)
Diabetes medications	179 (55.1)	1143 (57.2)	89 (61.4)	123 (45.7)	<.01	1534 (56.0)
Semaglutide	26 (8.0)	164 (8.2)	6 (4.1)	14 (5.2)	.13	210 (7.7)

ALT, alanine aminotransferase; AP, alkaline phosphatase; AST, aspartate aminotransferase; CT, computed tomography; MASH, metabolic dysfunction-associated steatohepatitis; MRI, magnetic resonance imaging; NASH, nonalcoholic steatohepatitis; VCTE, vibration-controlled transient elastography.

[a]Non-Hispanic Other includes patients without reported racial and ethnic group data; [b]Chi-square or Fisher exact test comparing categorical variables or Kruskal-Wallis comparing distribution of continuous variables; [c]Laboratory assessments summarized are those obtained closest to the index +/- 6 months; [d]Most recent diagnostic test results within 12 months prior to the index date.

- The highest Fibrosis-4 Index (FIB-4) scores were observed in non-Hispanic White patients and Hispanic/Latino patients (**Table 3**)

Table 3: Noninvasive tests at index stratified by racial and ethnic group among patients with non-cirrhotic MASH or cirrhosis enrolled in TARGET-NASH

Noninvasive test	Hispanic/ Latino n=325	Non-Hispanic White n=1999	Non-Hispanic Black n=145	Non-Hispanic Other[a] n=269	*P*[b]
FIB-4 score					<.01
n	229	1327	104	179	
Mean (SD)	2.8 (4.20)	3.1 (3.52)	1.9 (2.29)	2.4 (2.92)	
APRI score					<.01
n	229	1327	104	179	
Mean (SD)	1.1 (2.23)	1.0 (1.26)	0.9 (1.48)	0.9 (1.55)	
NFS score					<.01
n	267	1487	124	203	
Mean (SD)	-0.4 (2.40)	0.2 (2.31)	-0.2 (2.08)	-0.9 (2.36)	
FAST score					<.01
n	74	365	29	40	
Mean (SD)	0.4 (0.26)	0.5 (0.27)	0.4 (0.27)	0.4 (0.24)	

APRI, AST to platelet ratio; FAST, FibroScan–AST Score; FIB-4, Fibrosis-4 Index; MASH, metabolic dysfunction-associated steatohepatitis; NASH, nonalcoholic steatohepatitis; NFS, nonalcoholic fatty liver disease score.

[a]Non-Hispanic Other includes patients without reported racial and ethnic group data; [b]Chi-square or Fisher exact test comparing categorical variables or Kruskal-Wallis comparing distribution of continuous variables.

- The likelihood of progressing to all-cause mortality was lowest in non-Hispanic Black patients (IR, 0.32) and highest among non-Hispanic White patients (IR, 1.72; **Table 4**)

Table 4: All-cause mortality stratified by racial and ethnic group among patients with non-cirrhotic MASH or cirrhosis enrolled in TARGET-NASH

	Hispanic/ Latino n=325	Non-Hispanic White n=1999	Non-Hispanic Black n=145	Non-Hispanic Other[a] n=269
Incident new cases after index date[b]	29	206	3	11
Incidence proportion[c]	8.9	10.3	2.1	4.1
Mean person-time[d] (SD), months	59.7 (20.63)	59.7 (22.16)	64.0 (21.14)	60.5 (20.75)
Incidence rate, 100PM	1.50	1.72	0.32	0.68

MASH, metabolic dysfunction-associated steatohepatitis; NASH, nonalcoholic steatohepatitis; PM, person-months.

[a]Non-Hispanic Other includes patients without reported racial and ethnic group data; [b]Incident new cases is defined as the number of new events that occur between the day after index date and the earliest of the date of discontinuation or date of death or date of last contact; [c]Incidence proportion is defined as the number of new events that occur between the day after index date and the earliest of the date of discontinuation or date of death or date of last contact/(number of participants with a date of index date * 100); [d]A participant is eligible to contribute person-time only for the time during which he/she has not yet experienced the outcome and is therefore still at risk of developing the outcome. For participants experiencing the progression outcome: person-time is defined as the time (in months) from index date to the first time the outcome is observed. For participants not experiencing the progression outcome, person-time is defined as the time from index date to the earliest of the date of discontinuation or date of death or date of last known contact.

Conclusion

Significant differences in demographic and clinical characteristics, laboratory assessments, insurance type, and diagnostic testing were observed across racial and ethnic groups among patients enrolled in TARGET-NASH with non-cirrhotic MASH or cirrhosis with non-Hispanic White patients having the greatest incidence of all-cause mortality

References:
(**1**) Younossi et al. Global epidemiology of nonalcoholic fatty liver disease-meta-analytic assessment of prevalence, incidence, and outcomes. *Hepatology.* 2016;64(1):73-84. (**2**) Rich NE et al. Racial and ethnic disparities in nonalcoholic fatty liver disease prevalence, severity, and outcomes in the United States: a systematic review and meta-analysis. *Clin Gastroenterol Hepatol.* 2018;16(2): 198-210. (**3**) Barritt AS et al. Patient determinants for histologic diagnosis of NAFLD in the real world: a TARGET-NASH study. *Hepatol Commun*. 2021;5(6):938-946.

TARGET RWE
BETTER EVIDENCE • BETTER HEALTH

This analysis was sponsored by Novo Nordisk. The authors acknowledge the medical writing assistance of Victoria Jeter, PhD, of Precision AQ. ARM, BM, and HLM are employees of Target RWE. AS and RL are employees of Novo Nordisk Inc.

Presented at the American Association for the Study of Liver Diseases The Liver Meeting®; November 15-19, 2024; San Diego, California.