Time to Disease Progression and All-Cause Mortality in Adult MASH Patients in Real-World US Cohort

A. Sidney Barritt IV[1], Philip N. Newsome[2], Heather L. Morris[3], Feng Yu[3], Andrea R. Mospan[3], Taruja Karmarkar[4], Matthew G. Melaragno[4], Xiao Zhang[4], Mickie H. Cheng[4], Gail Fernandes[4], Arun J. Sanyal[5], on behalf of the TARGET-NASH Investigators
[1]University of North Carolina, Chapel Hill, NC, USA; [2]University of Birmingham, Birmingham, UK [3]Target RWE, Durham, NC, USA; [4]Merck & Co., Inc., Rahway, NJ, US; [5]Virginia Commonwealth University, Richmond, VA, USA

TARGET RWE
BETTER EVIDENCE • BETTER HEALTH

Background

- Metabolic dysfunction-associated steatohepatitis (MASH) burden is increasing, with approximately one-third of MASL patients progressing into MASH[1]
- Time to disease progression and incidence rates of liver related events outside of clinical trials and in real world clinical practice is unclear

Objective:

- To assess the time to disease progression for patients with MASH without cirrhosis, MASH with compensated cirrhosis and MASH with decompensated cirrhosis in a real-world setting

Methods:

Study Design and Data Source:

- A retrospective cohort study was conducted using the TARGET-NASH database - a real-world longitudinal observational cohort following >6000 patients enrolled across academic and community sites in the US
- Patients are enrolled in TARGET-NASH upon a diagnosis of MASLD by a treating physician in routine care
- Data was used from 08/01/2016 through 01/26/2024 (study period)
- Baseline demographics and clinical characteristics were assessed in the 36 months prior to the index date (defined as the date of enrollment in TARGET-NASH)
- Eligible patients were grouped into three subgroups for this study:
 - MASH without cirrhosis (MASH)
 - MASH with compensated cirrhosis (CC)
 - MASH with decompensated cirrhosis (DCC)

Statistical Analysis:

- Time-to-event analysis[2] from enrollment was used to assess the following outcomes: 1) time to MASH with compensated cirrhosis, 2) time to the first decompensation event in patients with compensated cirrhosis and 3) time to all-cause mortality in each of the 3 sub-cohorts

References: [1] Singh, S., et al., *Fibrosis progression in nonalcoholic fatty liver vs nonalcoholic steatohepatitis: a systematic review and meta-analysis of paired-biopsy studies.* Clinical gastroenterology and hepatology, 2015. **13**(4): p. 643-654. e9. [2] Kim HT. Cumulative incidence in competing risks data and competing risks regression analysis. *Clin Cancer Res* 2007;13:559-565.

Figure 1. Cumulative Incidence of Disease Progression and All-Cause Mortality

a. MASH progression to cirrhosis

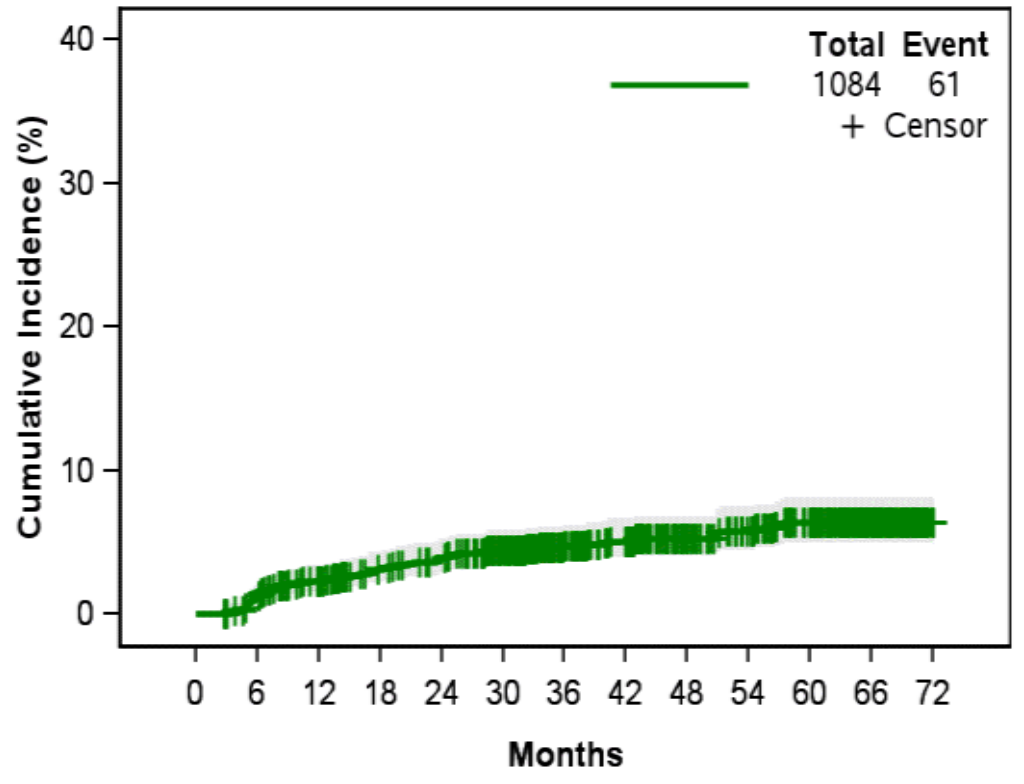
Total Event
1084 61
+ Censor
Cumulative Incidence (%)
Months

b. Compensated cirrhosis progression to decompensated cirrhosis

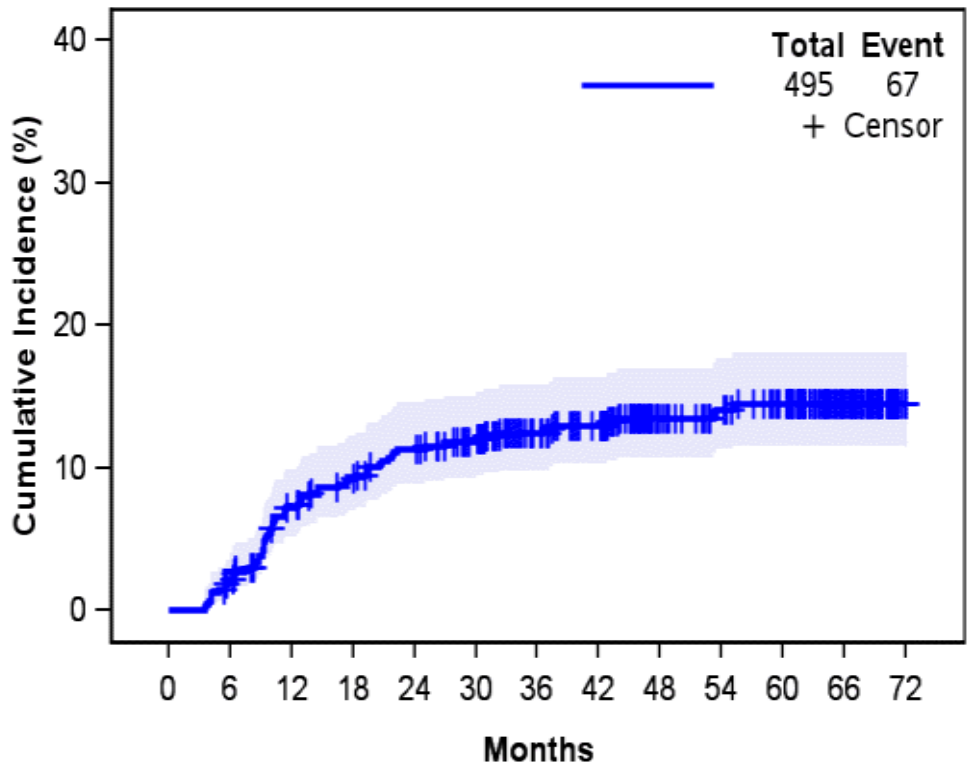
Total Event
495 67
+ Censor
Cumulative Incidence (%)
Months

c. All-cause mortality

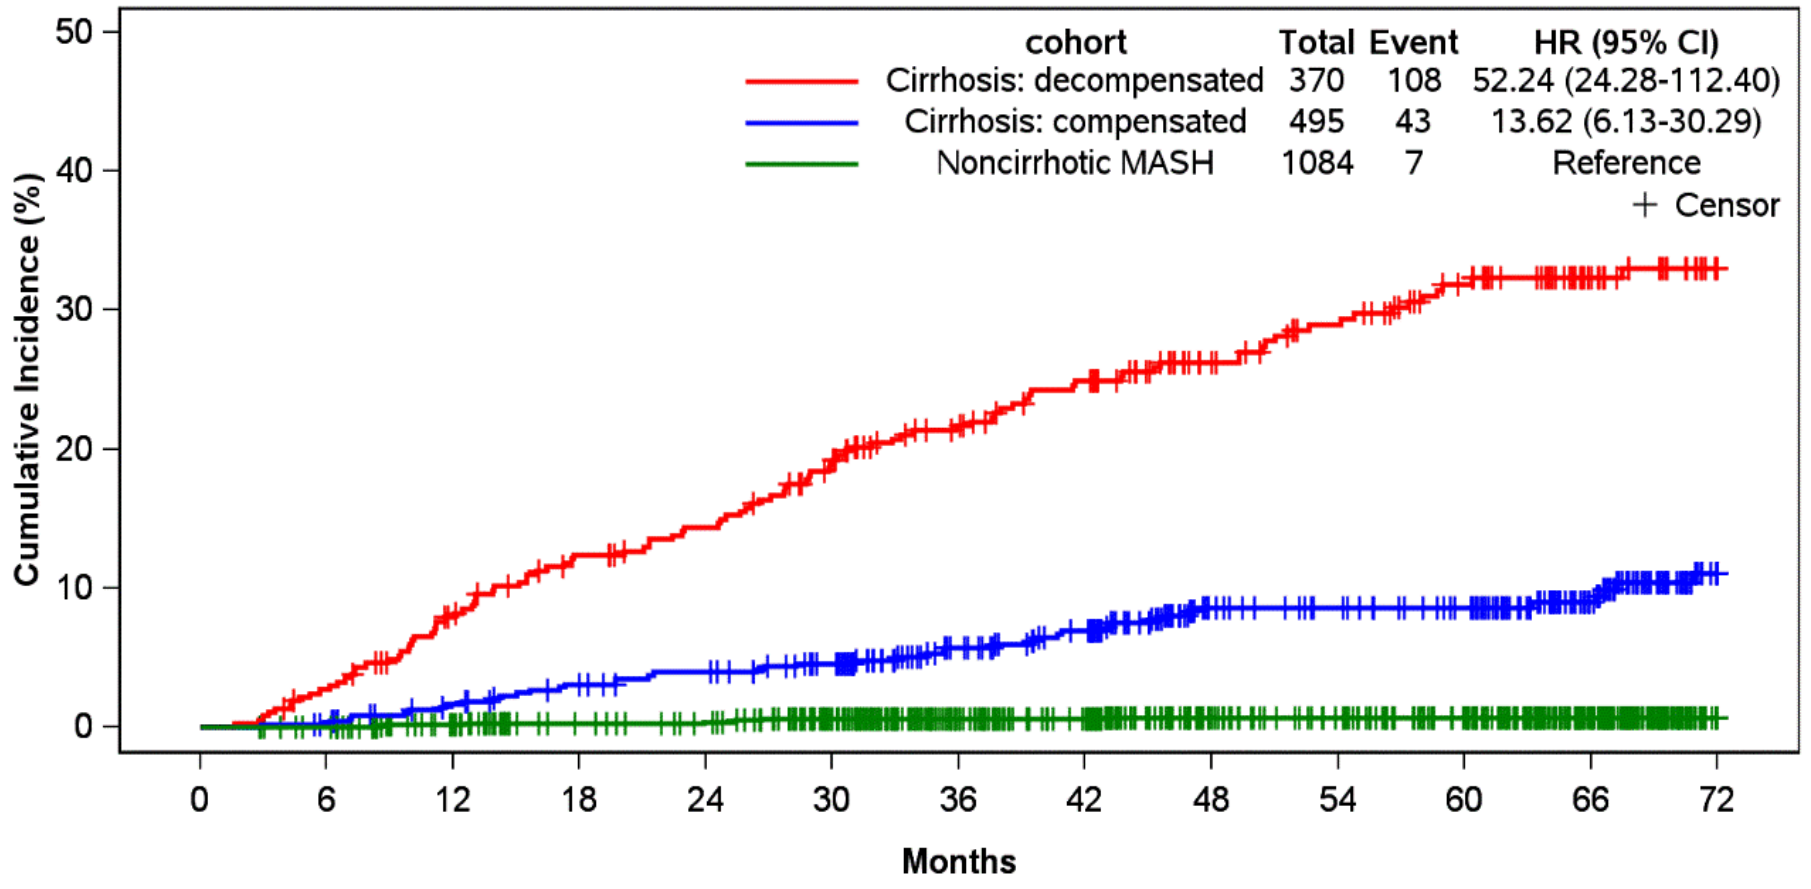
cohort Total Event HR (95% CI)
Cirrhosis: decompensated 370 108 52.24 (24.28-112.40)
Cirrhosis: compensated 495 43 13.62 (6.13-30.29)
Noncirrhotic MASH 1084 7 Reference
+ Censor
Cumulative Incidence (%)
Months

Note: Patients were censored at the time of follow-up if they did not have the event of interest. CIF=Cumulative Incidence Function

Results

- 1,949 participants enrolled in TARGET-NASH met the inclusion criteria for the analysis and were categorized into 3 subgroups (Table 1)

Table 1. Patient Demographics at Enrollment

	MASH (n=1084)	Compensated Cirrhosis (n=495)	Decompensated Cirrhosis (n=370)
Median Age (years)	55	62	63
Female, n(%)	689 (64)	297 (60)	205 (55)
Non-Hispanic White, n(%)	720 (66)	387 (78)	303 (82)
Academic Setting, n(%)	688 (64)	387 (78)	304 (82)
Private Insurance, n(%)	717 (66)	250 (51)	145 (39)
FIB-4 category[1], n(%)			
Low	588 (66)	82 (19)	14 (4)
Indeterminate	254 (28)	132 (30)	60 (17)
High	56 (6)	227 (52)	290 (80)
Hypertension, n(%)	659 (61)	397 (80)	352 (95)
Type 2 Diabetes, n(%)	496 (46)	321 (65)	266 (72)
Obesity, n(%)	698 (64)	357 (72)	269 (73)
CYP3A Inhibitors, n(%)	106 (10)	67 (14)	66 (18)

- Incidence rates of MASH progression to CC and compensated to DCC were 1.39 and 3.53 per 100-person year, respectively
- Among patients with MASH, 6% progressed to compensated cirrhosis within 72 months (Figure 1a)
- Among patients with CC, 14% had a decompensation event within 72 months (Figure 1b)
- Incidence rates for mortality were 0.14, 2.04, and 8.45 per 100-person year for MASH, CC and DCC, respectively
- MASH patients with CC and DCC had ~10% and 30% risk of death within 72 months, respectively
- Patients with DCC were at an increased risk of all-cause mortality (HR 52.24, 95% CI 24.28, 112.40) followed by CC (HR 13.62, 95% CI 6.13, 20.39) compared to patients with MASH (Figure 1c)

Limitations: Real-world databases are subject to potential missingness, limited generalizability and surveillance bias.

Conclusions

- Overall, we observed an increasing risk of mortality with greater disease severity
- In a cohort of MASH patients followed in routine clinical practice, those with compensated cirrhosis had a 10% probability of mortality within 72 months
- These results demonstrate the need for new therapies for patients with MASH cirrhosis.

Acknowledgements and Disclosures: Target RWE communities are collaborations among academic & community investigators, the pharmaceutical industry and patient community advocates. Target RWE communities are sponsored by TARGET PharmaSolutions Inc (d.b.a., Target RWE). The authors would like to thank all the investigators, participants, and research staff associated with TARGET-NASH. ClinicalTrials.gov Identifier: NCT02815891. TK, MM, XZ, MC and GF are employees of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, and may own stock and/or hold stock options in Merck & Co., Inc., Rahway, NJ, USA. This project was funded by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA

AASLD 2024 • Abstract • November 15 – 19, 2024 • San Diego, CA